Abstract ID
464
NCT05267626

Determination of the phase 2 dose of AU-007, an AI-designed human monoclonal antibody that redirects IL-2 to T effector cells

Tim Wyant[1,2], Drew Rasco[3], Meredith McKean[4], Andrew Haydon[5], Andrew Weickhardt[6], Sophia Frentzas[7], Elizabeth Ahern[7], John Powderly[8], Paul De Souza[9], Vinod Ganju[10], Siwen Hu-Lieskovan[11], Ganessan Kichenadasse[12], Jenny Tang[1], Lori Richards[1], Aron Knickerbocker[1], Inbar Amit[2], Yanay Ofran[1,2], James Vasselli[1]

[1]Aulos Bioscience, Larkspur, CA; [2]Biolojic Design, Rehovot, Israel; [3]START Center for Cancer Care, San Antonio, TX; [4]Sarah Cannon Research Institute, Tennessee Oncology, Nashville, TN; [5]The Alfred Hospital, Melbourne, Australia; [6]Austin Health, Heidelberg, Australia; [7]Monash Health, School of Medical and Health Sciences, Monash University, Melbourne, Australia; [8]Carolina BioOncology Institute, Huntersville, NC; [9]University of Sydney, Sydney, Australia; [10]Peninsula and Southeast Oncology, Frankston, Australia; [11]Huntsman Cancer Institute, University of Utah, Salt Lake City, UT; [12]Southern Oncology Cancer Research Institute, Bedford Park, Australia

aulos

Introduction

- AU-007 is an antibody that binds to human interleukin-2 (IL-2), redirecting IL-2 away from cells expressing a trimeric receptor that includes the CD25 alpha subunit (regulatory T cells, vascular endothelial cells, and eosinophils), and toward CD8+ T and natural killer (NK) effector cells expressing a dimeric receptor lacking the CD25 subunit.
- AU-007 is unique in the IL-2 therapeutic class in its ability to reduce the regulatory T cell (Treg) count by binding free IL-2 (endogenous or exogenous) and redirecting it to the dimeric IL-2 receptor expressed on CD8+ T and NK effector cells, expanding those cell populations. This has the effect of biasing the immune system toward activation rather than suppression.
- The AU-007 + IL-2 clinical trial is a Phase 1/2 dose escalation and expansion trial. The Phase 1 trial of AU-007 has completed the dose escalation phase and moved into Phase 2 expansion cohorts in selected tumor types. Data from pharmacodynamic and pharmacokinetic evaluations, as well as clinical efficacy and safety, were used to determine the dose for Phase 2 expansion.

Methods

This Phase 1/2 trial was designed as a 3+3 safety escalation trial with expansion. During Phase 1 dose escalation, AU-007 was administered as a short, intravenous infusion and increased as either A) monotherapy, B) in combination with low-dose, subcutaneous aldesleukin given with the initial dose of AU-007, or C) in combination with low-dose, subcutaneous aldesleukin given with each dose of AU-007 (Figure 1). Within any particular AU-007 dose level, the subcutaneous aldesleukin was also escalated. Data from the dose escalation phase were used to identify the dose(s) used to test in the Phase 2 dose expansion portion of the trial. Results from safety assessments, pharmacokinetic (PK) assays, and pharmacodynamic assays were used to identify the dose. Pharmacokinetic data were derived using standard antibody PK techniques. Pharmacodynamic measures included changes in circulating cell populations and peripheral expression of interferon-gamma (IFN-γ). Circulating cells were determined by flow cytometry and peripheral blood cytokines were determined using electrochemiluminescence. Data from the assays were collated and compared across dosing regimens and levels to determine the dose used in the expansion. To assess the level of correlation between progression-free survival (PFS) and Treg reduction, the largest reduction achieved after Day 3 was calculated for each participant. Patients were then grouped by those who achieved less than the median Treg reduction and those who achieved more than the median Treg reduction, and PFS was plotted on a Kaplan-Meier curve.

Figure 1: Phase 1 Dose Escalation

Dosing began late Q2 2022

The Phase 1 dose escalation is complete, and no patients experienced dose-limiting toxicity (DLT). Once the DLT period cleared in an escalation cohort, additional "backfill" patients (up to a total of 10 in each escalation cohort) were allowed to enroll in each escalation cohort. 55 patients were enrolled into the three Dose Escalation Arms: Arm 1A: 15; Arm 1B: 12 (1 Backfill); Arm 1C: 28 (11 Backfill).

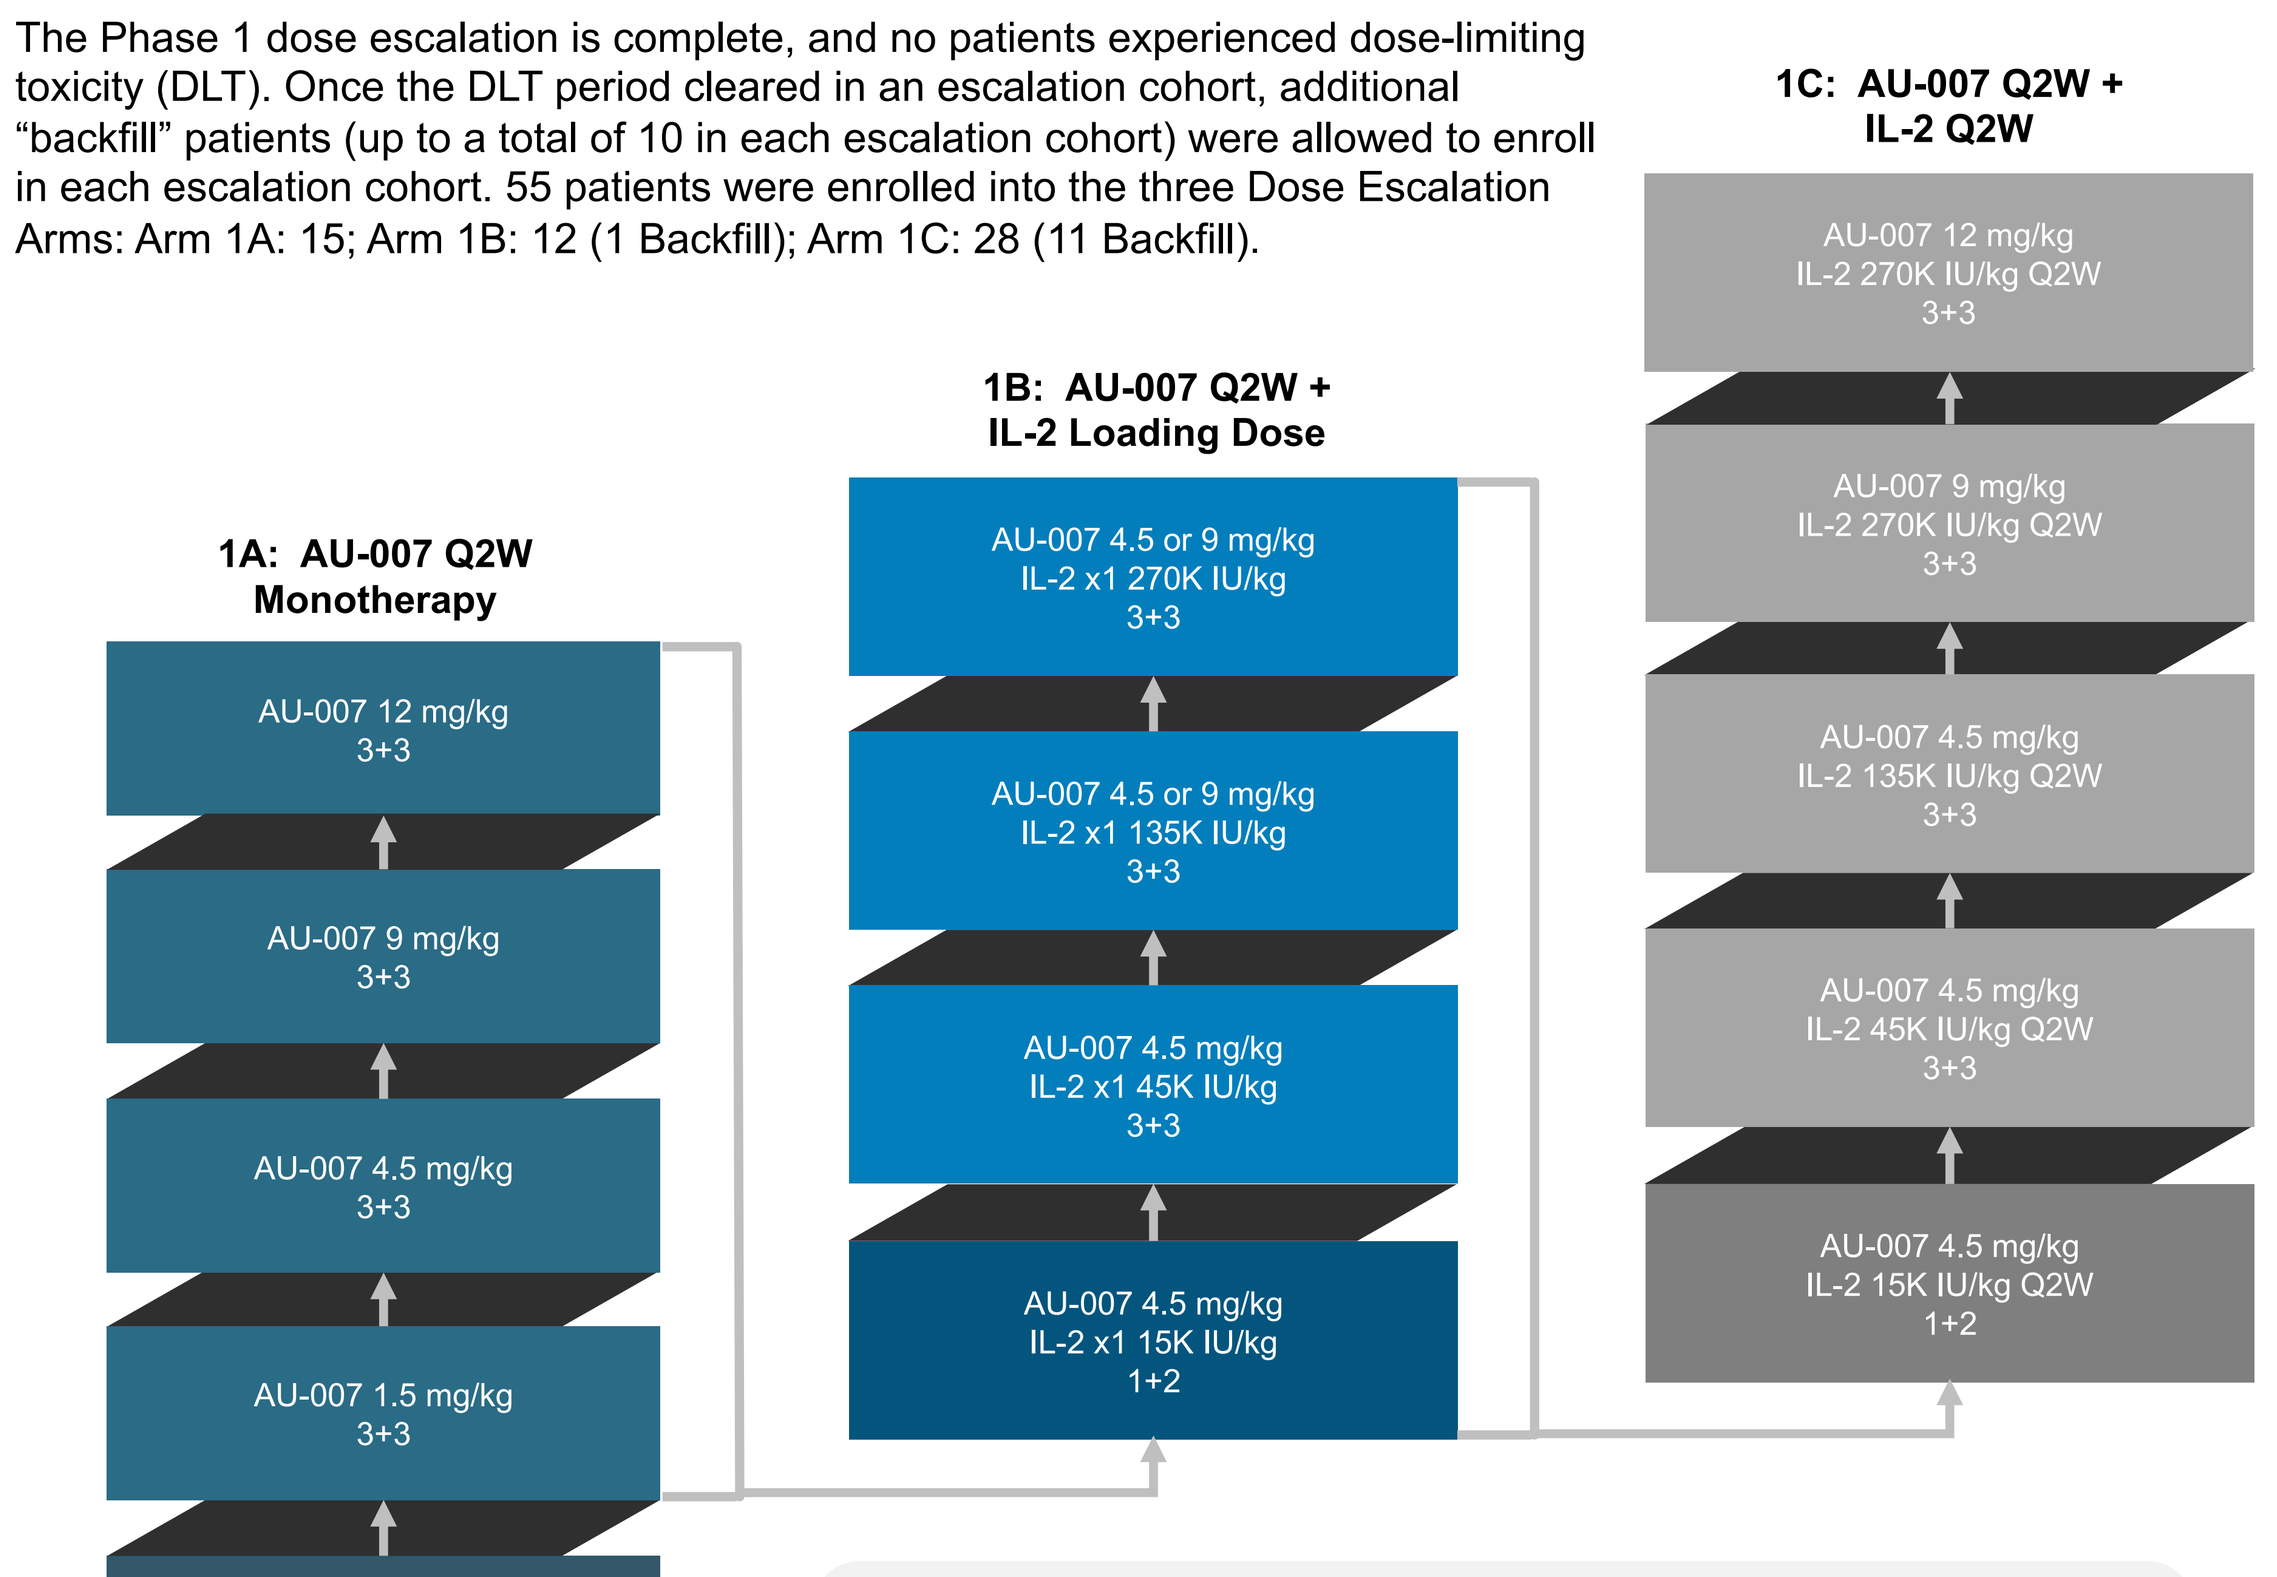
1A: AU-007 Q2W Monotherapy
AU-007 12 mg/kg 3+3
AU-007 9 mg/kg 3+3
AU-007 4.5 mg/kg 3+3
AU-007 1.5 mg/kg 3+3
AU-007 0.5 mg/kg 1+2
1B: AU-007 Q2W + IL-2 Loading Dose
AU-007 4.5 or 9 mg/kg IL-2 x1 270K IU/kg 3+3
AU-007 4.5 or 9 mg/kg IL-2 x1 135K IU/kg 3+3
AU-007 4.5 mg/kg IL-2 x1 45K IU/kg 3+3
AU-007 4.5 mg/kg IL-2 x1 15K IU/kg 1+2
1C: AU-007 Q2W + IL-2 Q2W
AU-007 12 mg/kg IL-2 270K IU/kg Q2W 3+3
AU-007 9 mg/kg IL-2 270K IU/kg Q2W 3+3
AU-007 4.5 mg/kg IL-2 135K IU/kg Q2W 3+3
AU-007 4.5 mg/kg IL-2 45K IU/kg Q2W 3+3
AU-007 4.5 mg/kg IL-2 15K IU/kg Q2W 1+2

In this trial, aldesleukin is administered subcutaneously, at much lower doses and much less frequently than the approved regimen (600K IU/kg every 8 hours for 14 administrations) of intravenously administered aldesleukin.

Figure 2: Phase 2 Dose Expansion

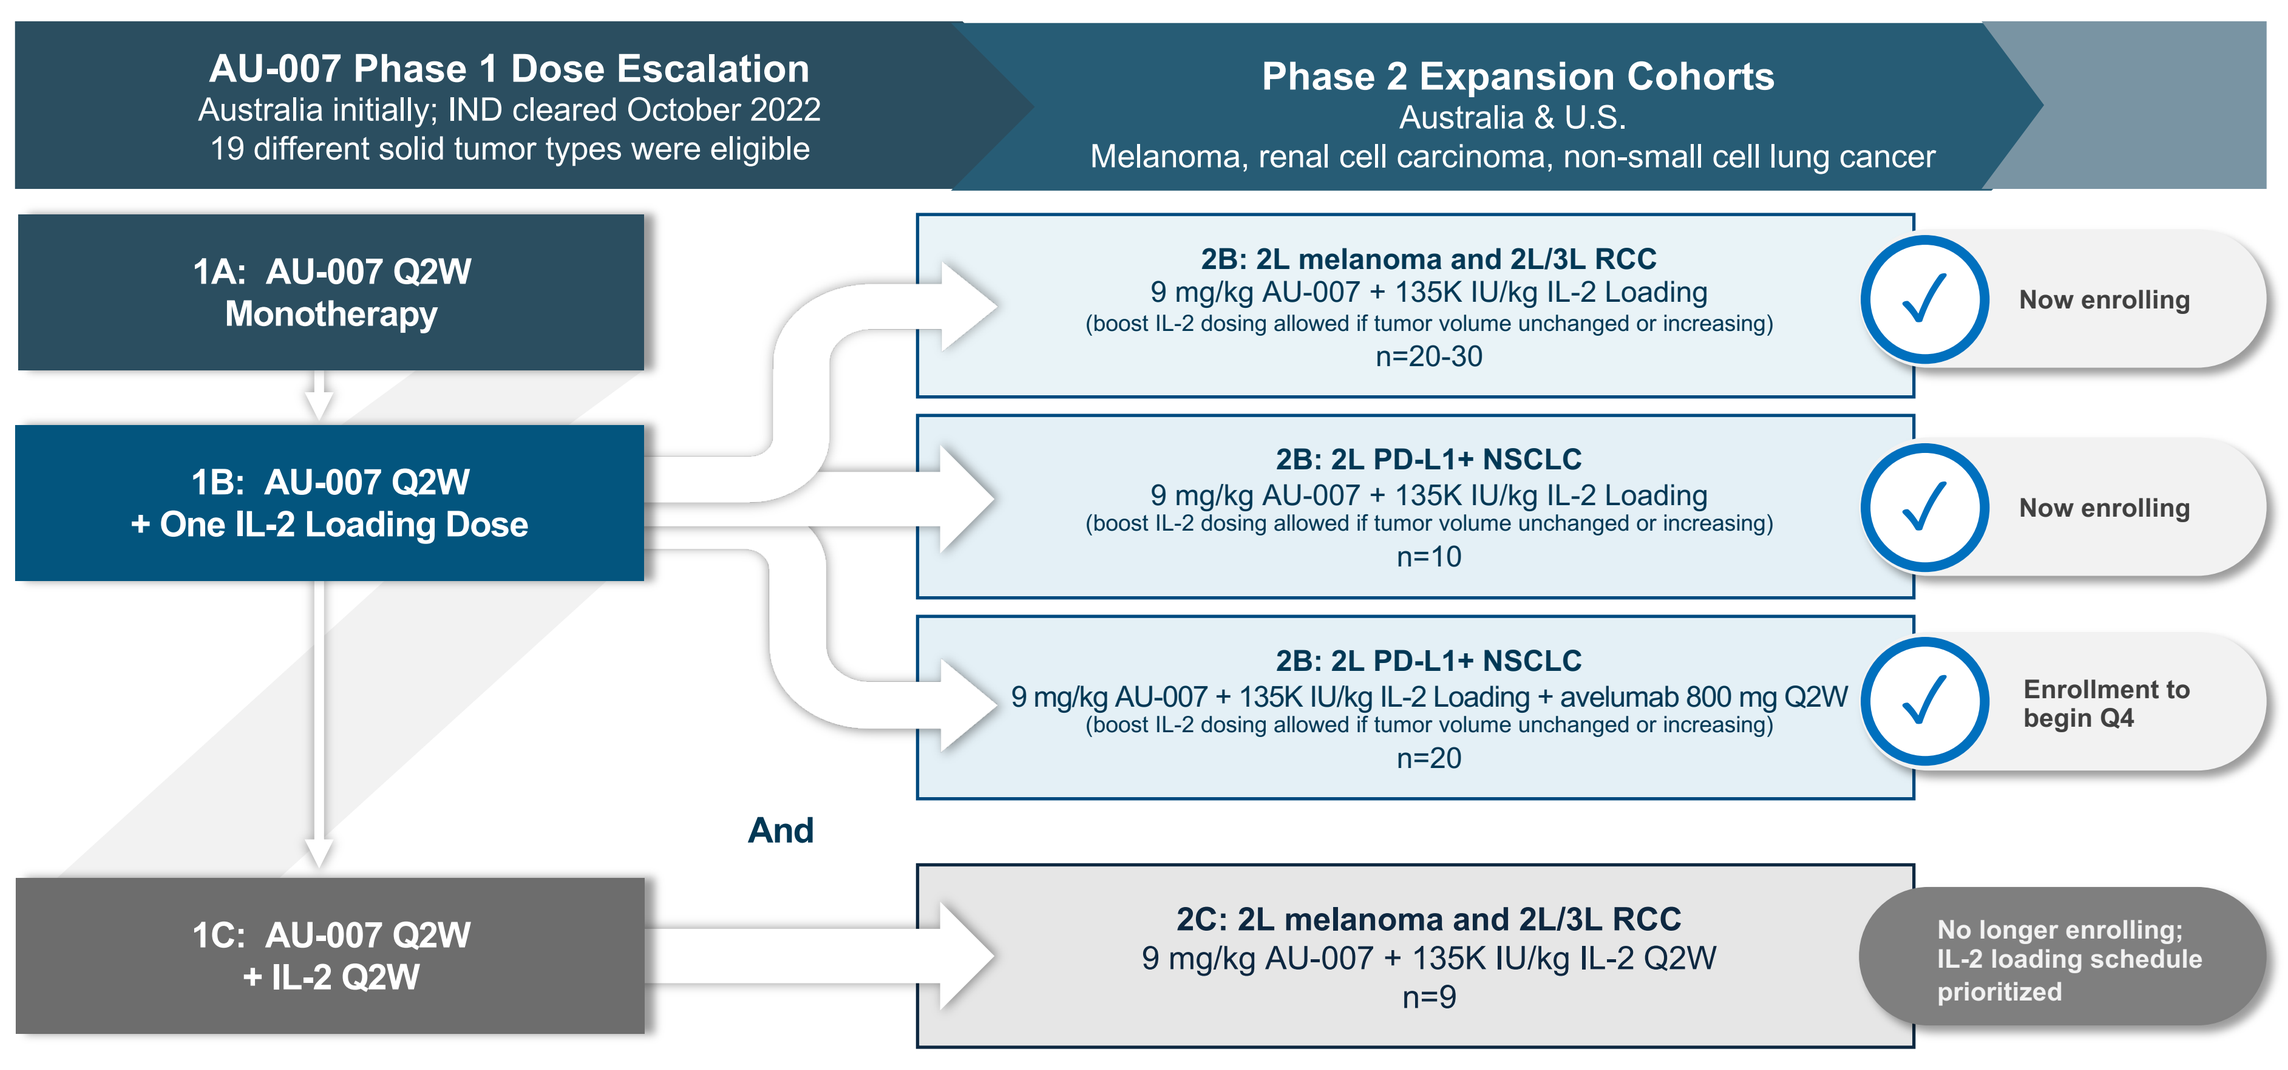
AU-007 Phase 1 Dose Escalation
Australia initially; IND cleared October 2022
19 different solid tumor types were eligible
Phase 2 Expansion Cohorts
Australia & U.S.
Melanoma, renal cell carcinoma, non-small cell lung cancer
1A: AU-007 Q2W Monotherapy
1B: AU-007 Q2W + One IL-2 Loading Dose
1C: AU-007 Q2W + IL-2 Q2W
2B: 2L melanoma and 2L/3L RCC
9 mg/kg AU-007 + 135K IU/kg IL-2 Loading
(boost IL-2 dosing allowed if tumor volume unchanged or increasing)
n=20-30
Now enrolling
2B: 2L PD-L1+ NSCLC
9 mg/kg AU-007 + 135K IU/kg IL-2 Loading
(boost IL-2 dosing allowed if tumor volume unchanged or increasing)
n=10
Now enrolling
2B: 2L PD-L1+ NSCLC
9 mg/kg AU-007 + 135K IU/kg IL-2 Loading + avelumab 800 mg Q2W
(boost IL-2 dosing allowed if tumor volume unchanged or increasing)
n=20
Enrollment to begin Q4
And
2C: 2L melanoma and 2L/3L RCC
9 mg/kg AU-007 + 135K IU/kg IL-2 Q2W
n=9
No longer enrolling; IL-2 loading schedule prioritized

Table 1: AU-007 Exhibits a PK Profile With Characteristics Typical of an IgG1-LALA Monoclonal Antibody With an Approximate 15-Day Half-Life in Humans

Pharmacokinetics				
Arm	AU-007 Dose (mg/kg)	N	Cmax (μg/mL)	AUClast (d*μg/mL)
1A	0.5	2	10.8 (16)	53.7 (105)
1A	1.5	3	29.6 (13)	231 (12)
1A	4.5	3	110 (15)	828 (42)
1A	9	4	255 (21)	1,700 (22)
1A	12	3	282 (9.1)	1,910 (39)
1B	4.5	12	132 (47)	639 (39)
1C	4.5	21	155 (42)	773 (46)
1C	9	4	223 (12)	1,350 (25)

- Overall, AU-007 PK approximately dose-proportional over the dose range tested.
- T1/2 ~15 days by population PK analysis.
- In general, comparable AU-007 PK exposures occur when dosed alone or in combination with subcutaneous aldesleukin.
- No evidence of neutralizing anti-drug antibodies (ADAs).

Table 2: Mean Fold Change in Peripheral Cell Populations at Day 43 Independent of AU-007 Dose (Group B Only)

Mean fold change on Day 43 in effector T cells and Tregs

IL-2 Dose Level	N=	CD8 Cells	NK Cells	Treg Cells	CD8/Treg Ratio
15K IU/kg	1	0.86	0.56	0.42	2.05
45K IU/kg	4	0.76	0.82	0.45	1.45
135K IU/kg	2	1.61	2.11	0.81	1.97
270K IU/kg	2	0.99	1.31	0.56	1.95

Cell populations were measured in peripheral blood samples by flow cytometry. Fold change was determined relative to the Day 1 pre-dose sample.

Figure 3: After a Single Dose of AU-007 and Low-Dose, Subcutaneous Aldesleukin, Fold Changes in Peripheral Populations at Day 15 Peak at 135K IU/kg Aldesleukin

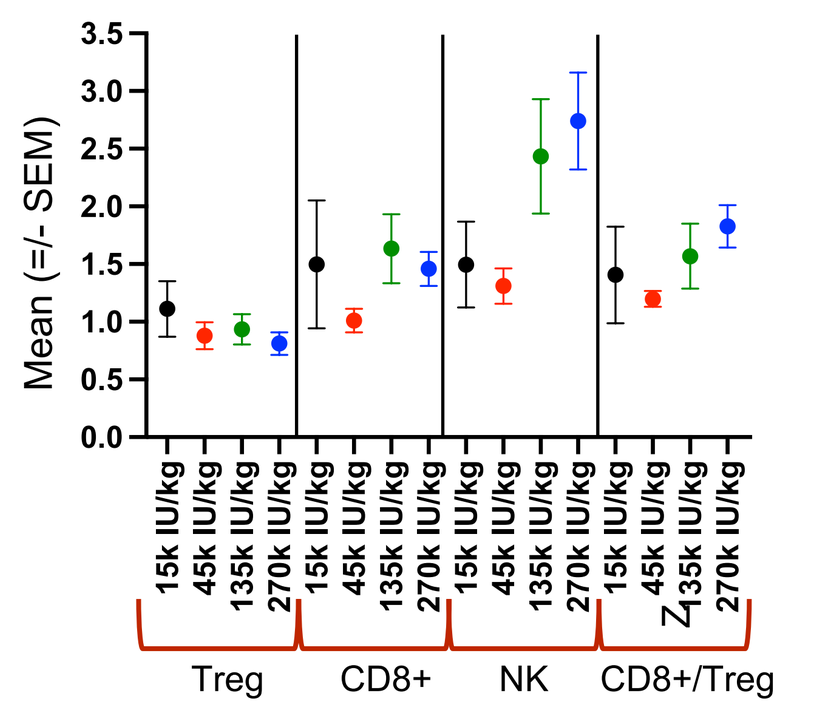
Mean (=/- SEM)
15k IU/kg
45k IU/kg
135k IU/kg
270k IU/kg
Treg
CD8+
NK
CD8+/Treg

Figure 3: Changes in the Day 15 levels of Treg, NK, CD8+ cell populations, and the CD8/Treg ratio across all cohorts dosed with any AU-007 and one of the following: 15K IU/kg, 45K IU/kg, 135K IU/kg, or 270K IU/kg subcutaneous aldesleukin. At Day 15, the Treg population did not change or was decreased relative to baseline. Ascending doses of subcutaneous aldesleukin tended to increase CD8+ T cells and NK cells and favored a higher CD8/Treg ratio.

Figure 4: Comparing Single to Multidose, the Fold Change From Baseline in Peripheral Cell Populations at Day 43 of Cycle 1 Also Peaks at 135K IU/kg Low-Dose, Subcutaneous Aldesleukin

AU-007 + low-dose, subcutaneous aldesleukin decreased circulating Tregs with escalating doses of subcutaneous aldesleukin increasing CD8+ T cells and NK cells. The CD8/Treg ratio increased in most patients in the escalation phase. AU-007 monotherapy has been previously shown to decrease circulating Treg cells (presented at ASCO 2024).

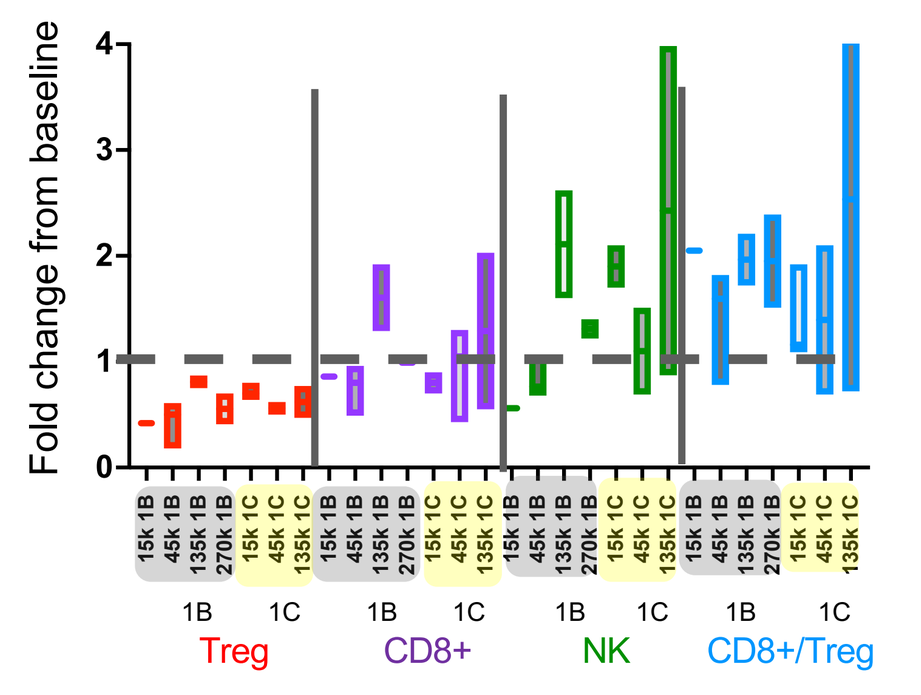
Fold change from baseline
1B
1C
Treg
CD8+
NK
CD8+/Treg

Figure 4: Changes in the Day 43 levels of Treg, NK, CD8+ cell populations, and the CD8/Treg ratio in 1B and 1C cohorts dosed with 4.5 mg/kg AU-007 and one of the following: 15K IU/kg, 45K IU/kg, 135K IU/kg subcutaneous aldesleukin (B and C), or 270K IU/kg for Cohort 1B. For all doses, the Treg population was decreased relative to baseline. Ascending doses of subcutaneous aldesleukin tended to increase CD8+ T cells and NK cells and favored a higher CD8/Treg ratio.

Figure 5: Average Percent Change in Tregs, CD8/Treg Ratio, and NK Cells in Phase 1 Showed Continuing Trends in Changes

Continuing trends in peripheral Treg cell decreases and CD8/Treg ratio and NK cell increases. NK cell increases appear more sustained in patients receiving Arm C dose regimen.

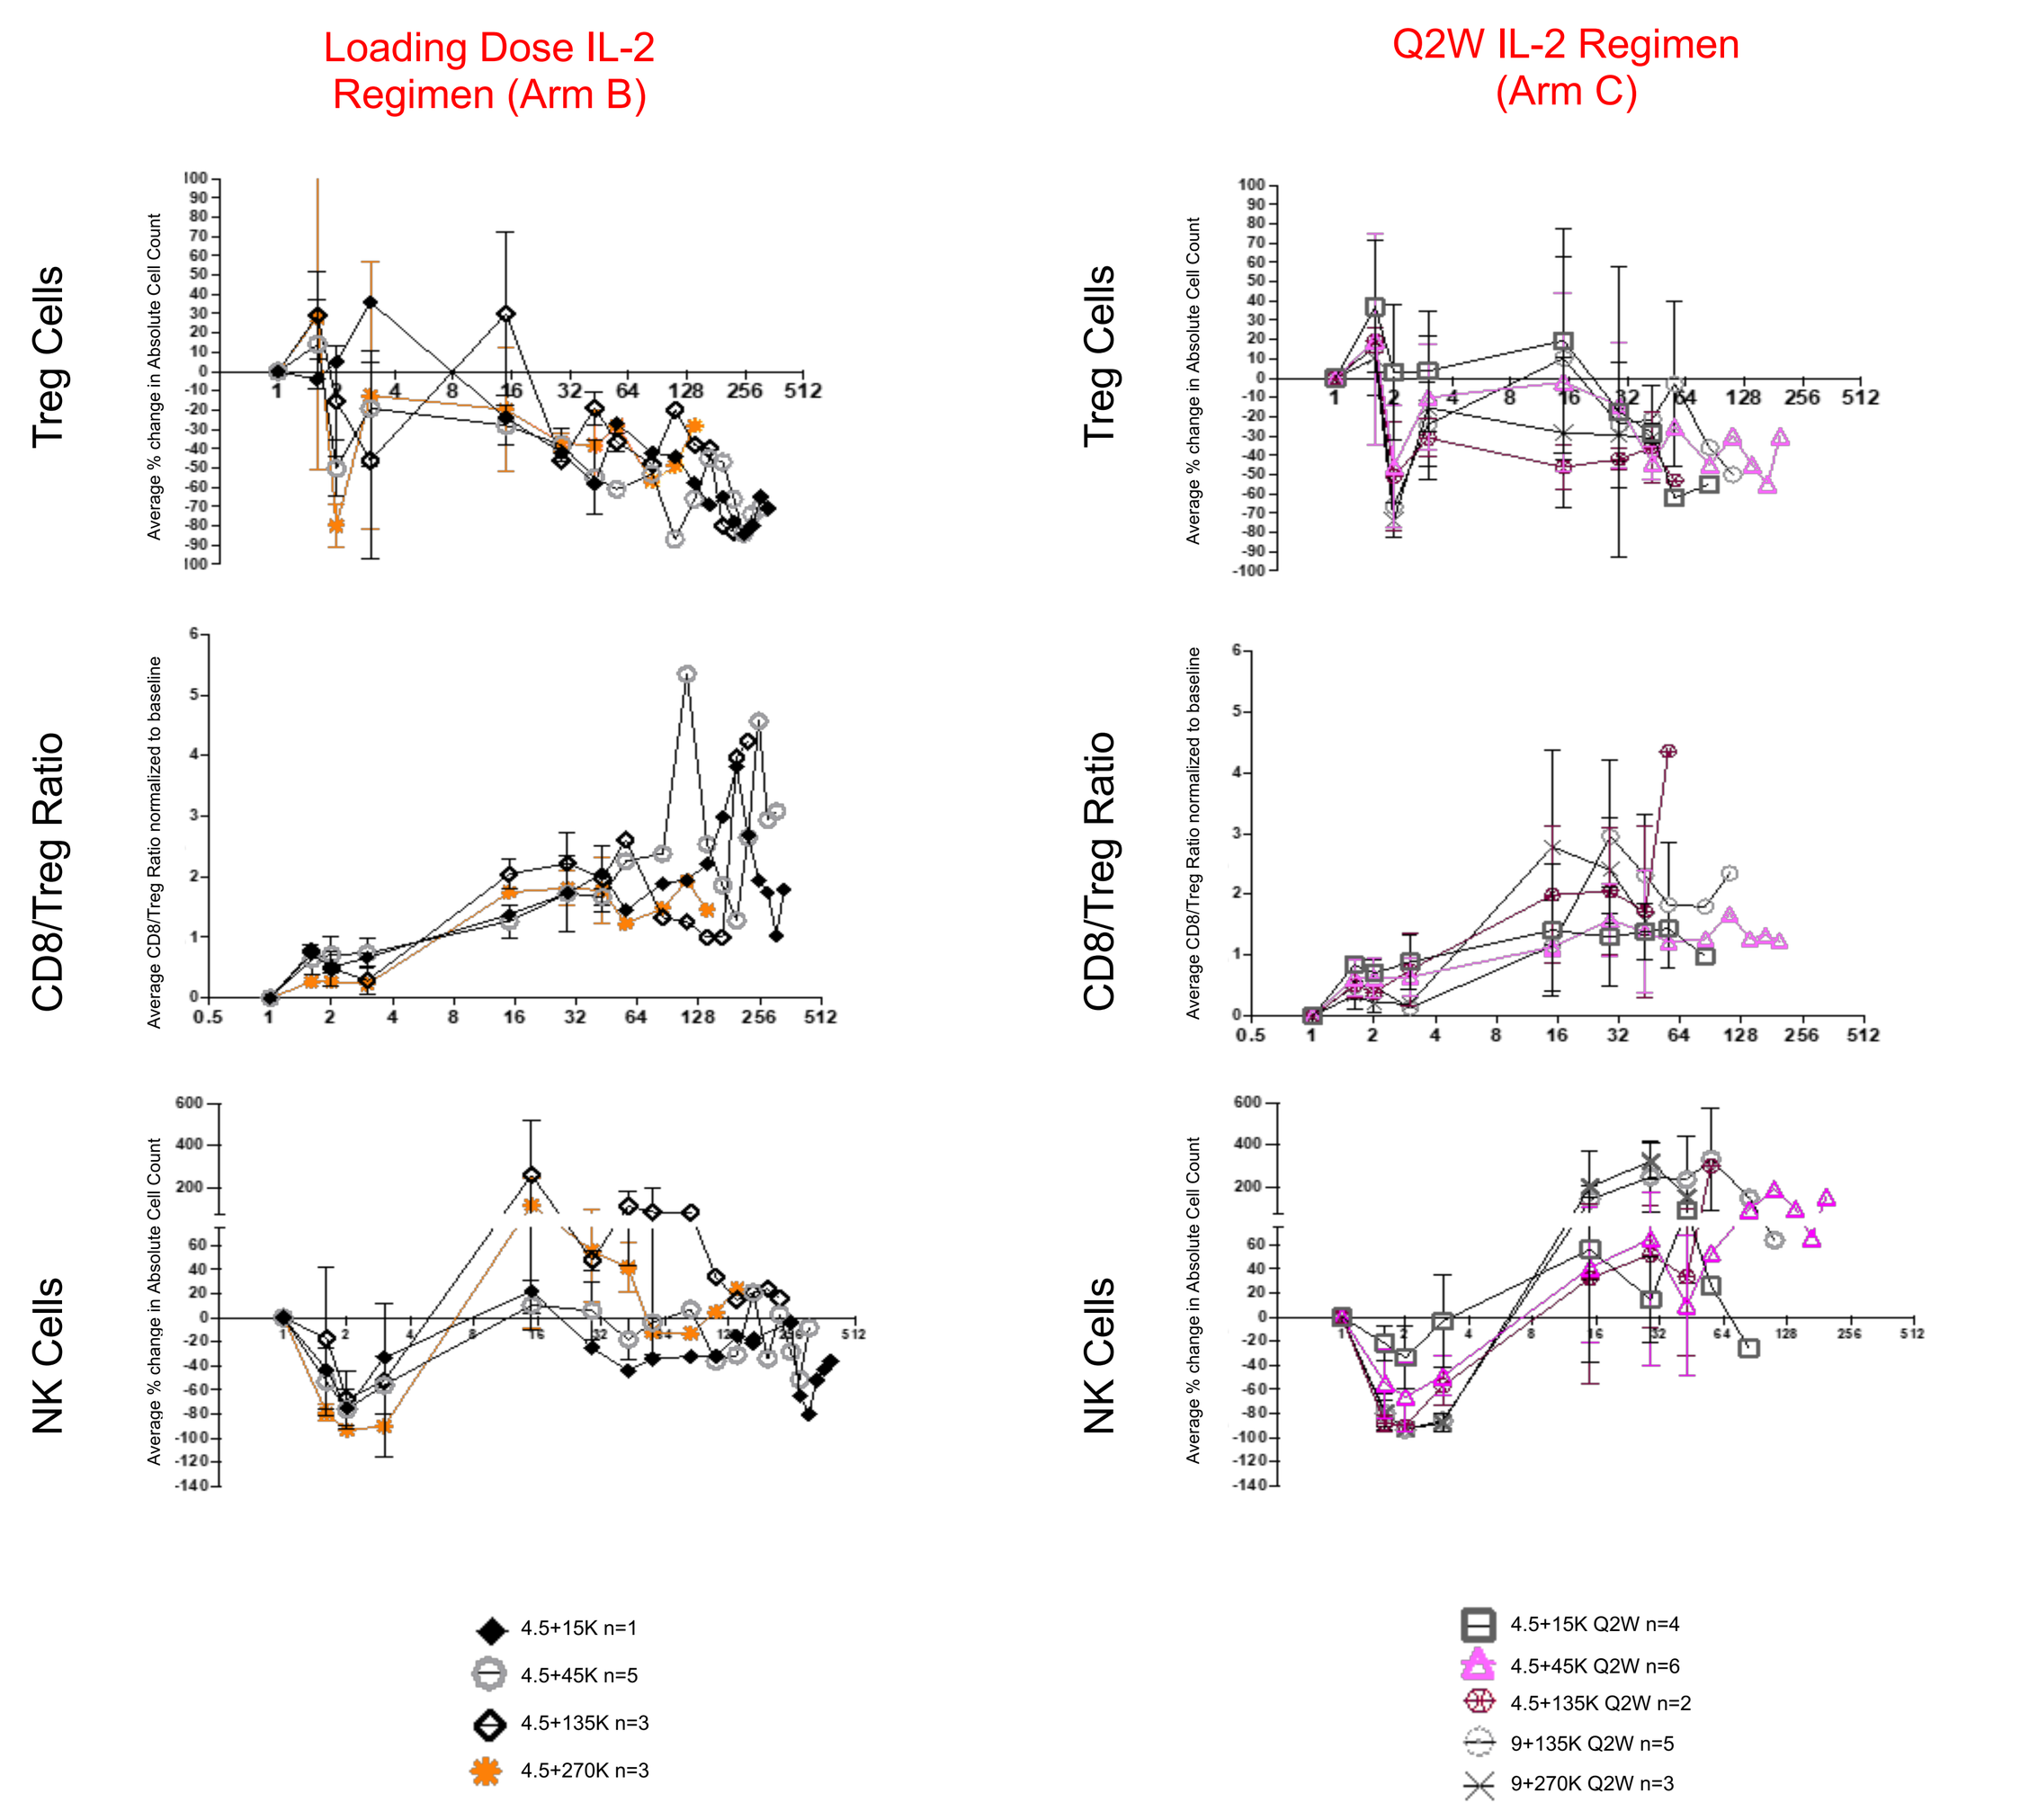
Loading Dose IL-2 Regimen (Arm B)
Q2W IL-2 Regimen (Arm C)
Treg Cells
CD8/Treg Ratio
NK Cells
Average % change in Absolute Cell Count
Average CD8/Treg Ratio normalized to baseline
4.5+15K n=1
4.5+45K n=5
4.5+135K n=3
4.5+270K n=3
4.5+15K Q2W n=4
4.5+45K Q2W n=6
4.5+135K Q2W n=2
9+135K Q2W n=5
9+270K Q2W n=3

Figure 6: Decreases (With Any IL-2 Schedule) in Tregs on Treatment Correlate With Longer PFS

Absolute cell count of Tregs per μl of blood: analysis based on median Treg reduction of 43%

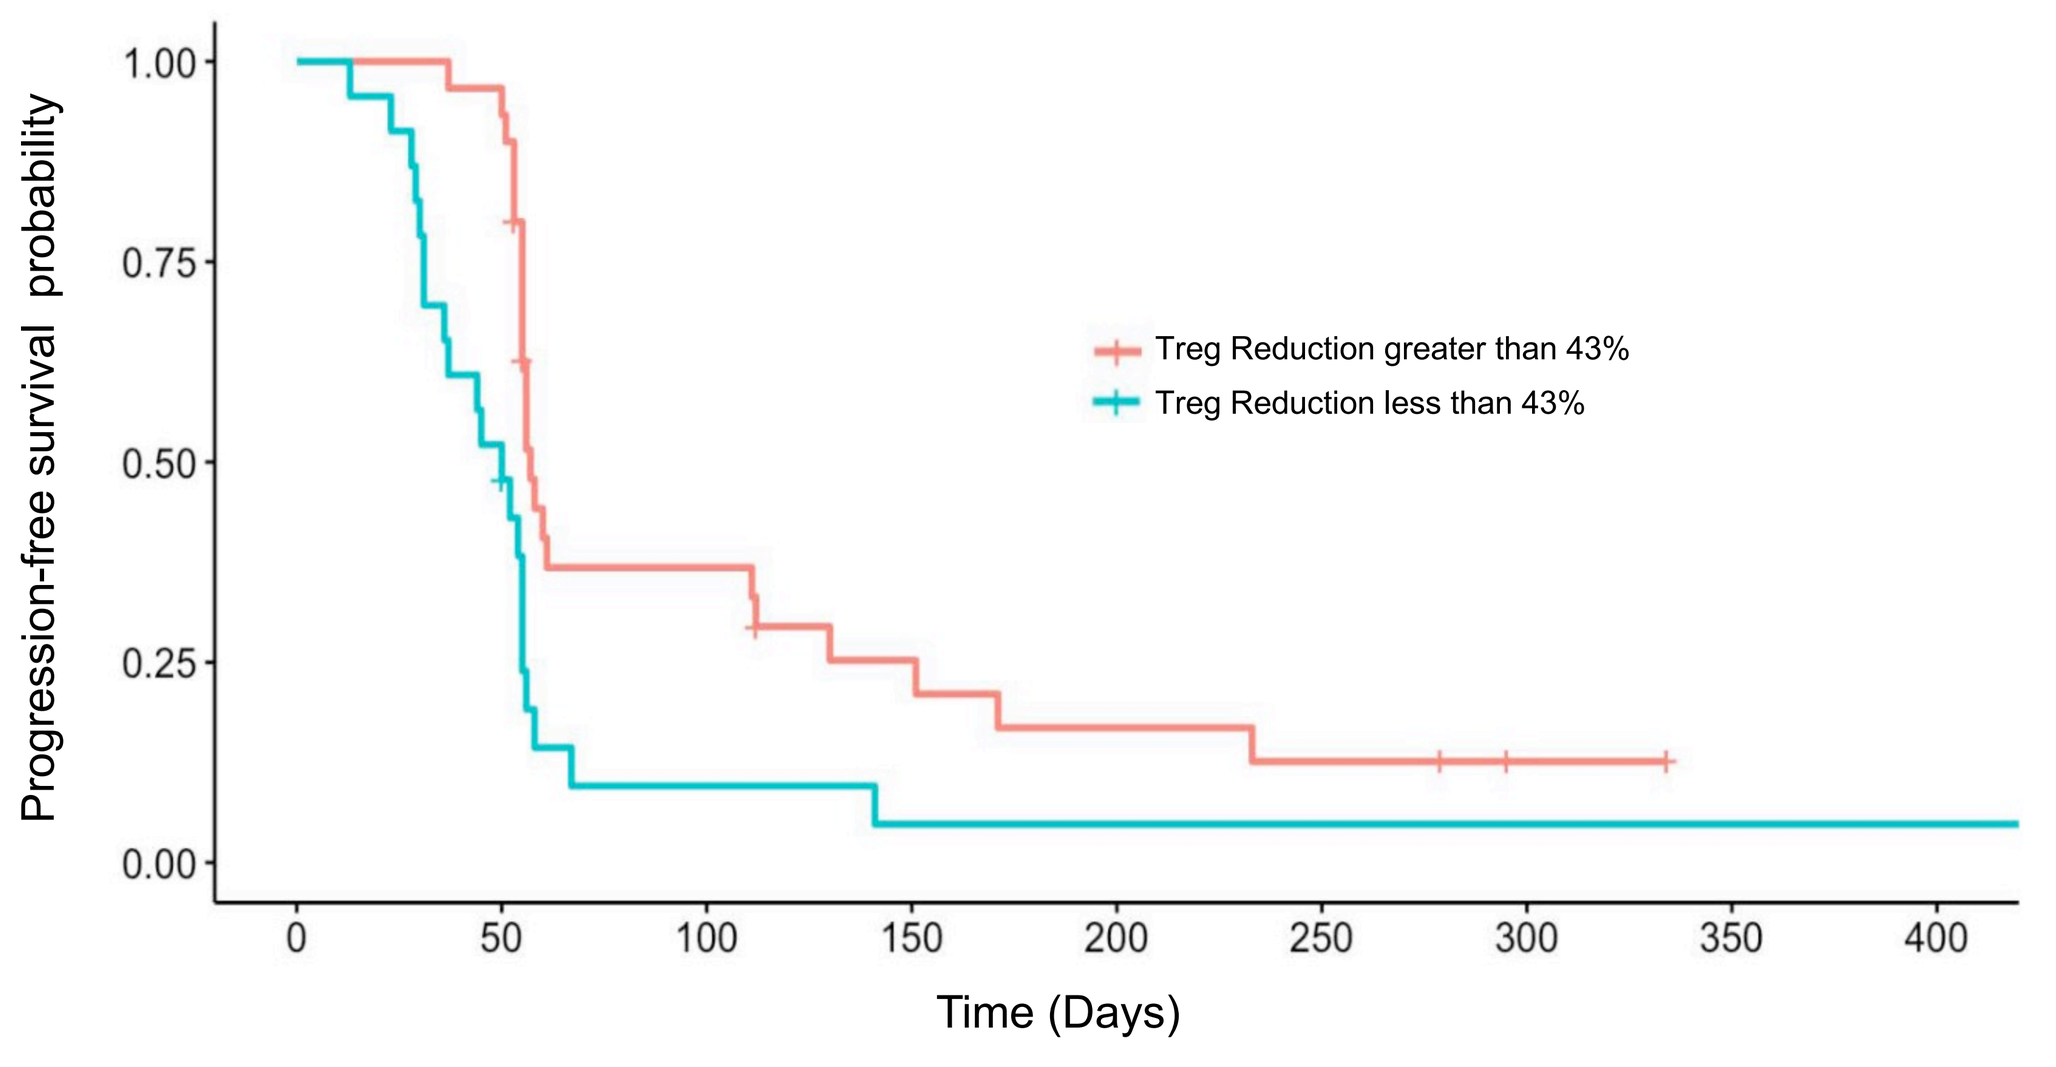
Progression-free survival probability
Time (Days)
Treg Reduction greater than 43%
Treg Reduction less than 43%

Results are calculated by deriving the median greatest Treg decrease across all patients (excluding the first 2 days on treatment) and dividing the patients who were ≤ the median or > than the median.

Figure 6: Larger decreases in circulating Treg cells were correlated with better PFS outcomes in patients from the Phase 1 and 2 portions of the trial, across a range of dose levels. Patients were grouped into those whose Treg reduction was less than or greater than the median reduction of 43%, and PFS was examined. Though numbers are small, the data suggest that larger decreases in Treg populations are associated with better clinical outcomes.

Figure 7: Dose Response Effect of IFN-γ and IL-2

There was an observed increase in the concentration of circulating IFN-γ as subcutaneous aldesleukin doses increased. No significant increase in peak IFN-γ was observed between the 135K IU/kg dose of subcutaneous aldesleukin and the 270K IU/kg dose.

Figure 7: Individual peak circulating IFN-γ levels within 2 days after dosing with AU-007 and subcutaneous aldesleukin. Ordinary one-way ANOVA was used to determine significance. Changes were observed across all AU-007 dose levels. Circulating IFN-γ rose with increasing doses of subcutaneous aldesleukin; however, no significant differences were observed between 135K IU/kg and 270K IU/kg of subcutaneous aldesleukin.

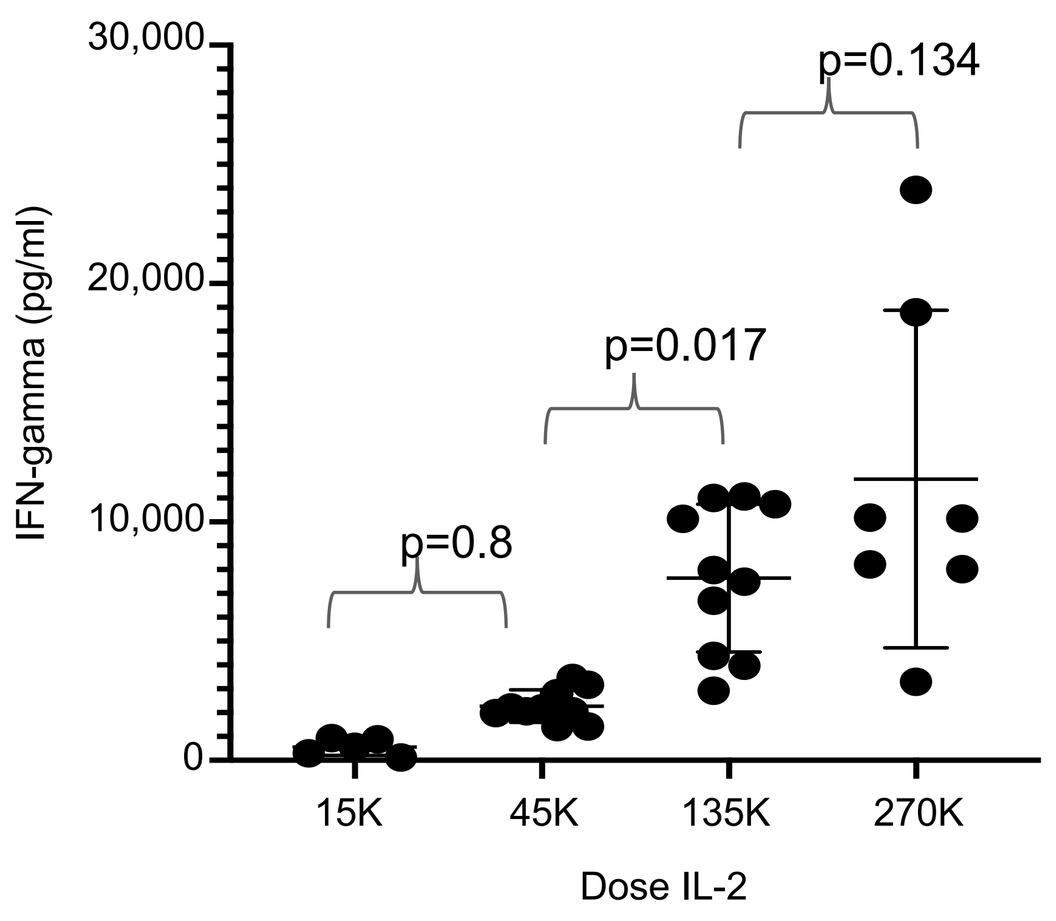
IFN-gamma (pg/ml)
p=0.8
p=0.017
p=0.134
15K
45K
135K
270K
Dose IL-2

Figure 8: 9 mg/kg AU-007 Enhances Early IFN-γ Production While Decreasing the Risk for Tumor Progression

Directly comparing 4.5mg/kg AU-007 + 135K IU/kg subcutaneous aldesleukin and 9 mg/kg AU-007 + 135K IU/kg subcutaneous aldesleukin, the 9 mg/kg AU-007 was associated with higher IFN-γ (Figure 4A) and was correlated with longer PFS (Figure 4B).

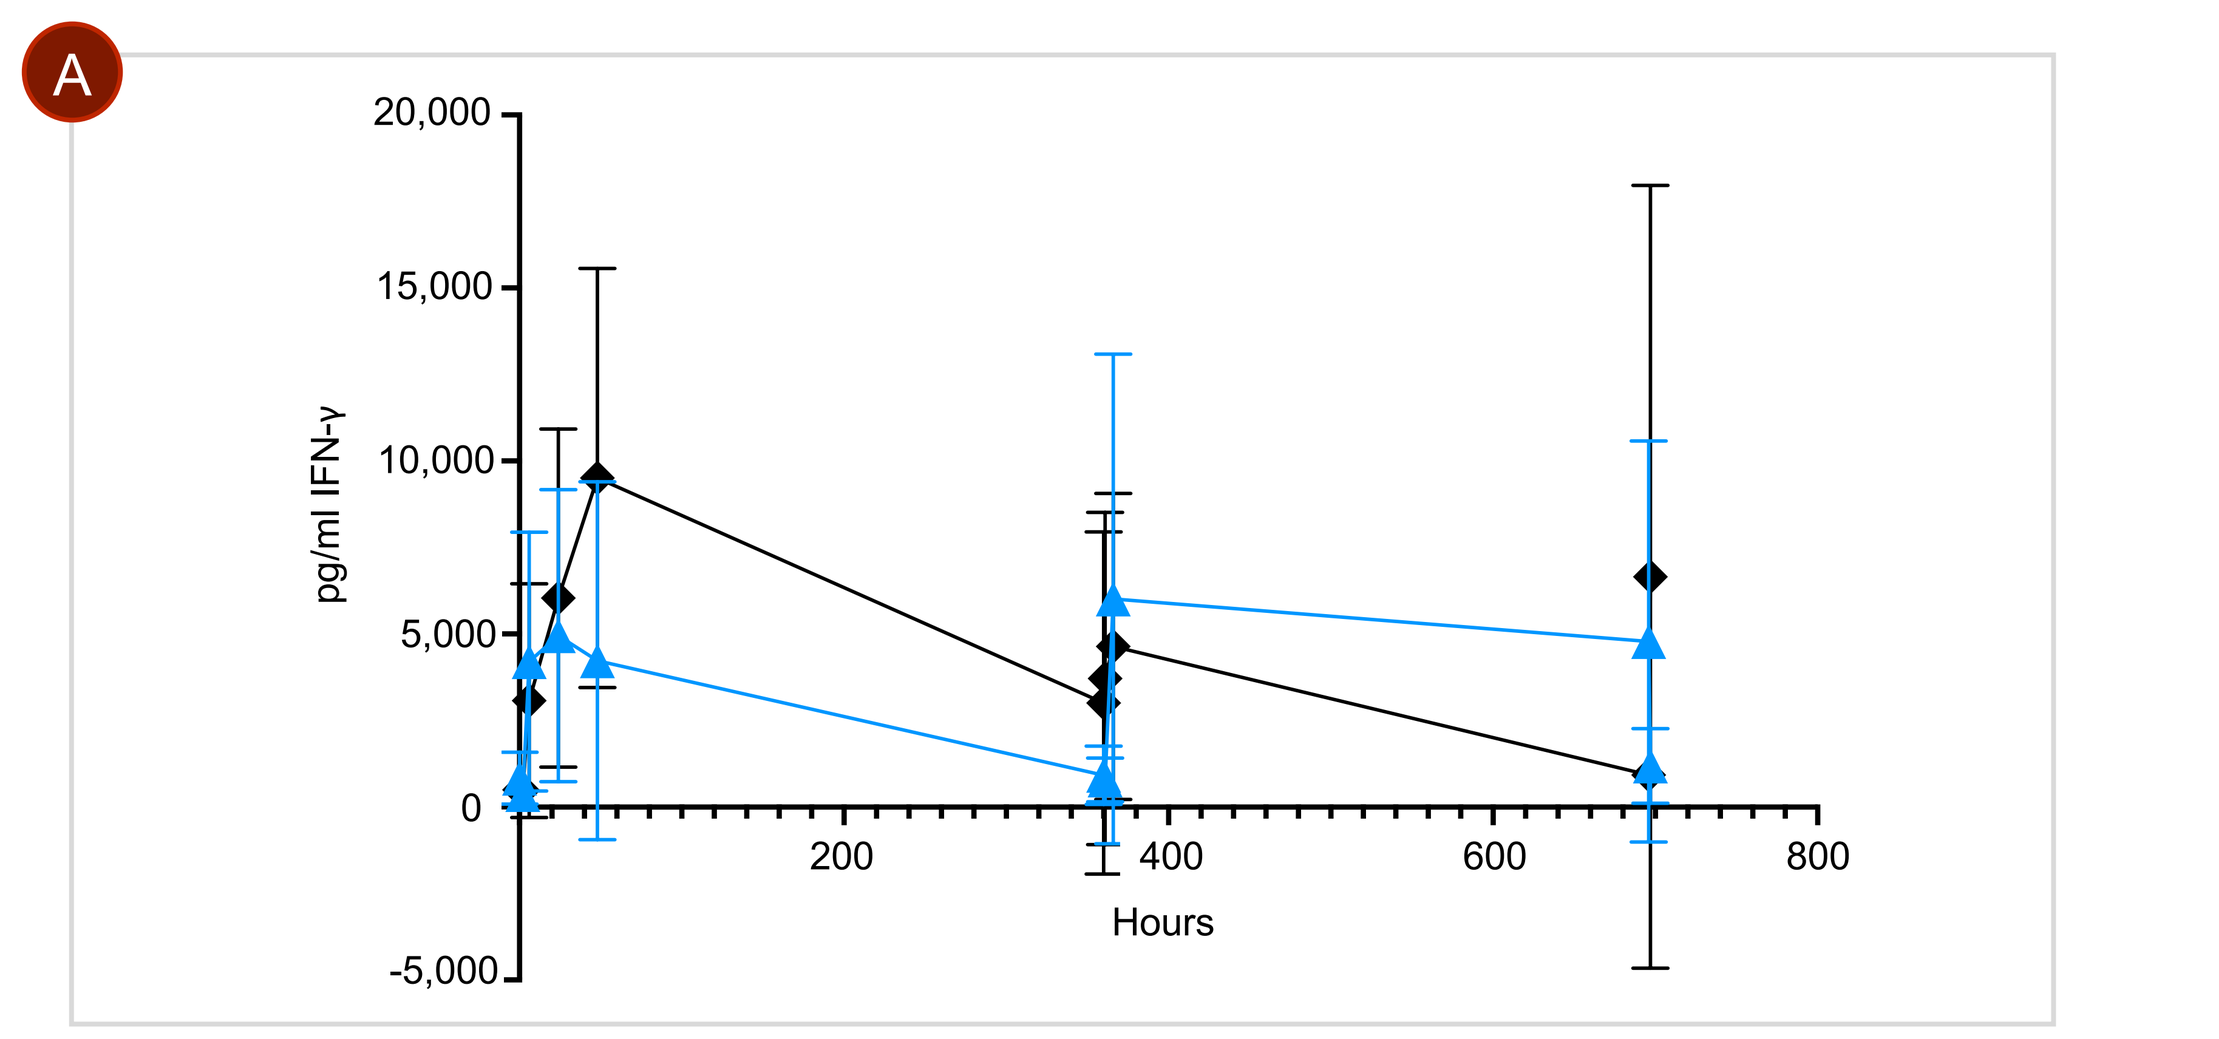
A
pg/ml IFN-γ
Hours

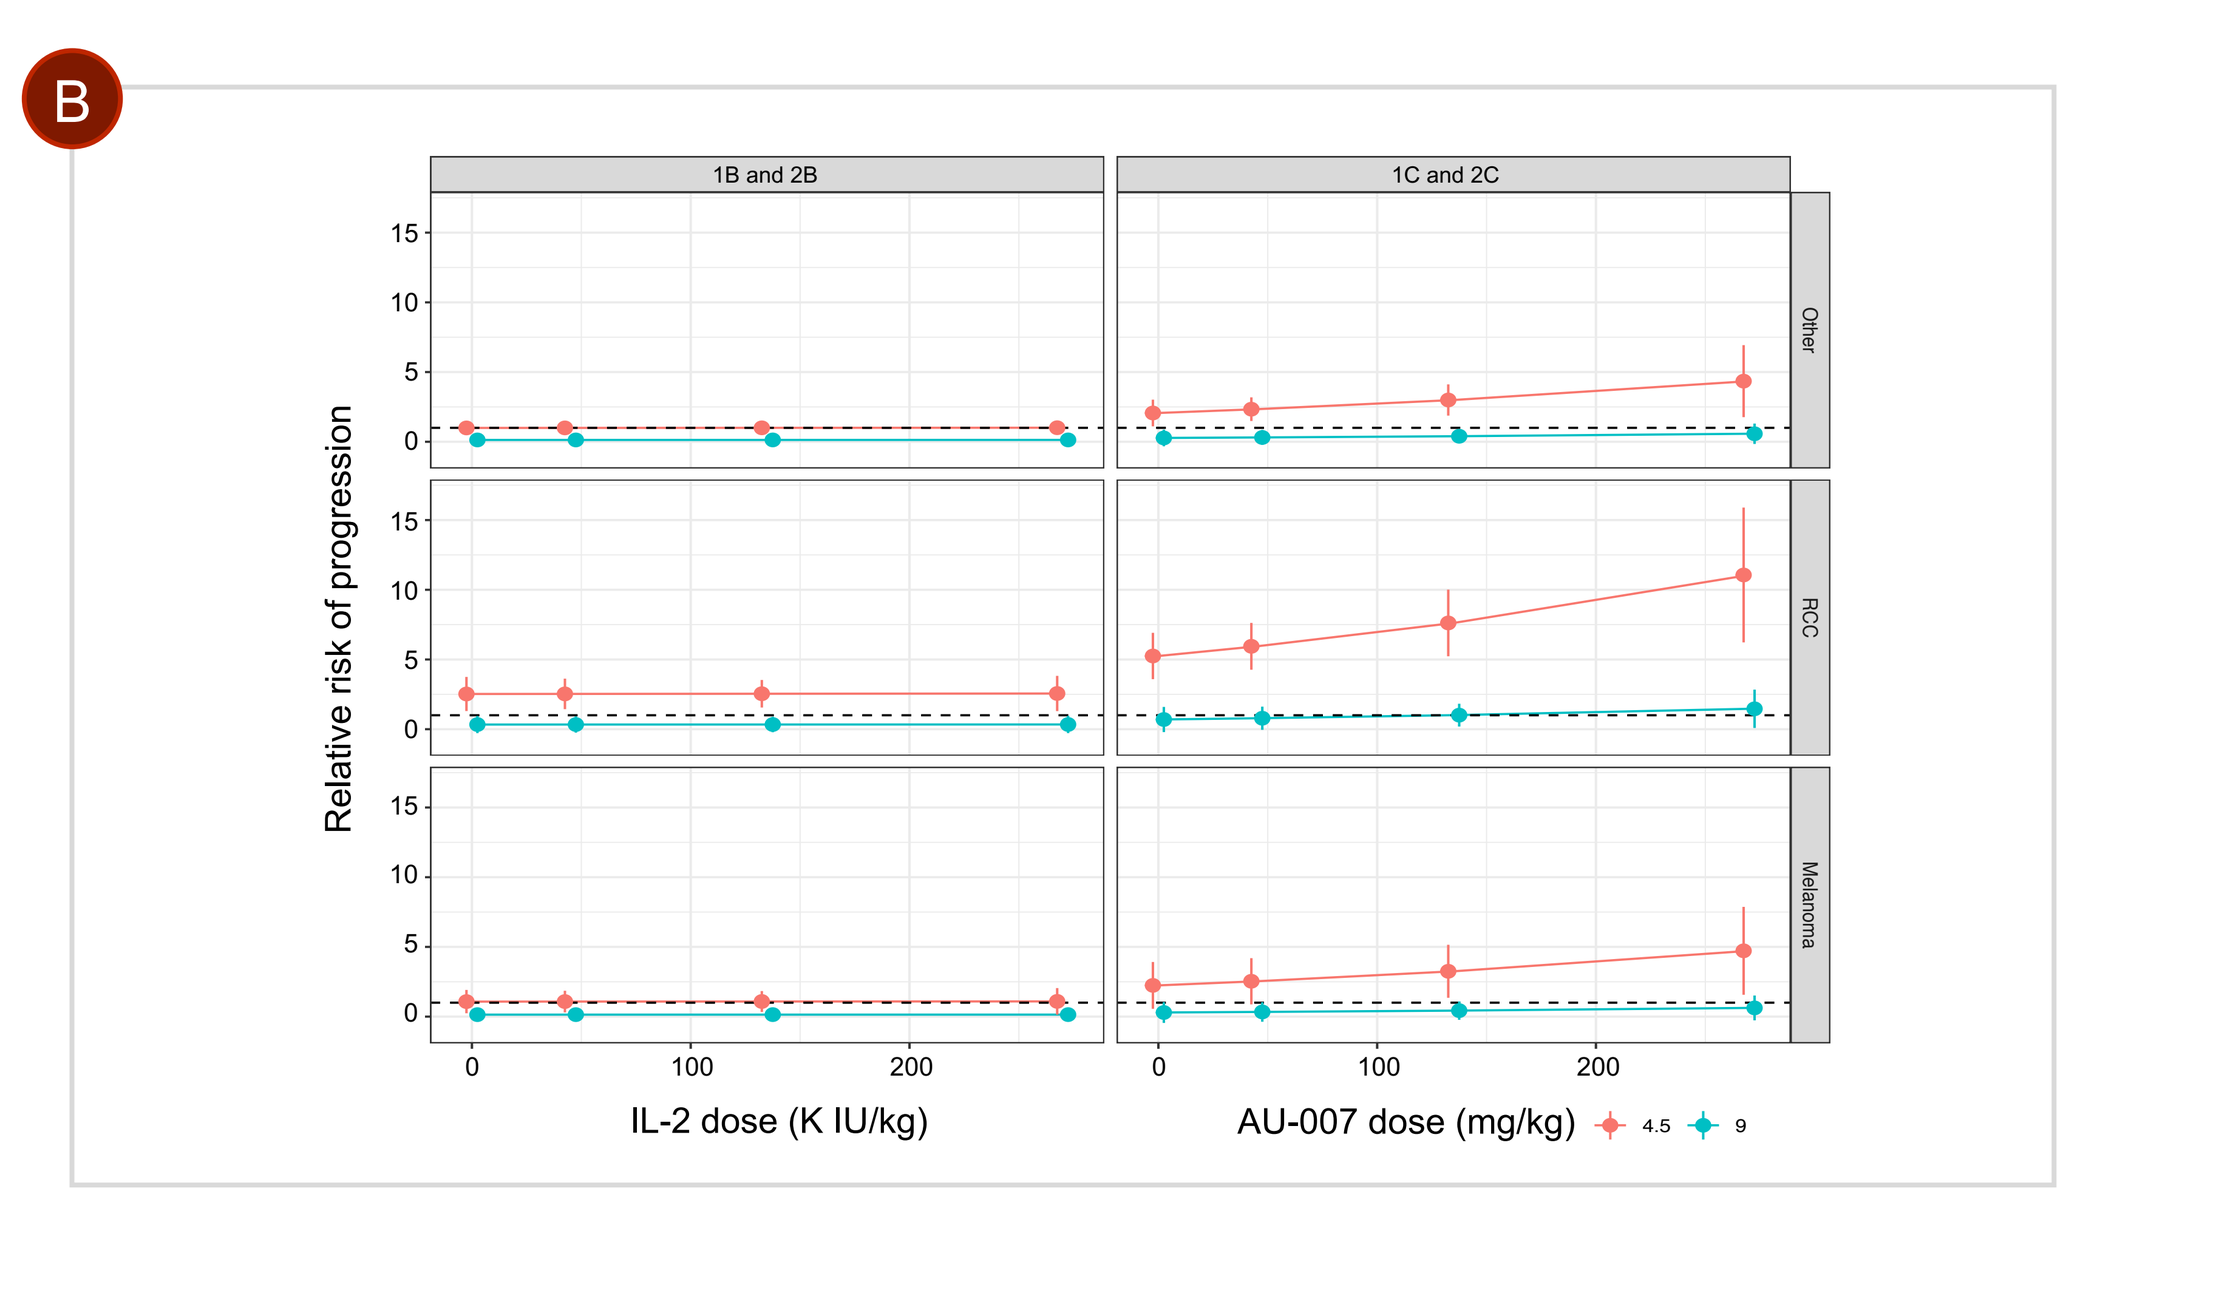
B
1B and 2B
1C and 2C
Other
RCC
Melanoma
Relative risk of progression
IL-2 dose (K IU/kg)
AU-007 dose (mg/kg)
4.5
9

Figure 8: Comparing 9 mg/kg AU-007 to 4.5 mg/kg AU-007 show dosing with 9 mg/kg is superior, as measured by IFN-γ levels achieved in the first 2 weeks, to dosing with 4.5 mg/kg. **A)** IFN-γ levels after dosing with 9 mg/kg AU-007 + 135K IU/kg of subcutaneous aldesleukin were higher in the first 2 weeks compared to dosing with 4.5 mg/kg AU-007 + 135K IU/kg of subcutaneous aldesleukin. **B)** Modeling of data from this clinical trial show that dosing with 9 mg/kg AU-007 is associated with a lower risk of progression compared to dosing with 4.5 mg/kg AU-007. This is most strikingly seen in the 1C and 2C cohorts that received subcutaneous aldesleukin every 2 weeks (Q2W).

Table 3: Safety Profile Remains Mild and Tolerable

Treatment with AU-007 monotherapy resulted in no drug-related serious adverse events (SAEs) or Grade 3/4 AEs. AU-007 + low-dose, subcutaneous aldesleukin demonstrated a manageable and tolerable safety profile.

	AU-007 Monotherapy	AU-007 + One IL-2 Dose	AU-007 + IL-2 Q2W	Total
Patients (n)	15	23	37	75
Any AE (%)	14 (93)	21 (91)	34 (92)	69 (92)
Drug-Related AEs (%)	4 (27)	18 (78)	25 (68)	47 (63)
Drug-Related SAEs (%)	0	3 (13)	2 (5)	5 (7)
Fever	0	1 (Gr2)	0	
Cytokine Release Syndrome (CRS)	0	1 (Gr2)	2 (Gr2, Gr4)	
Infusion-Related Reaction	0	1 (Gr2)	0	
Drug-Related Grade 3 or 4 AEs (%)	0	4 (17)	4 (11)[1]	8 (11)
Lymphopenia	0	3 (Gr4)	2 (Gr3, Gr4)	
CRS	0	0	1 (Gr4)	
Anemia	0	0	1 (Gr3)	
Increased Lipase	0	1 (Gr3)	0	
Dose-Limiting AEs (%)	0	0	1 (2.7)	1 (1.3)

1. One patient had 2 Gr3/4 AEs: lymphopenia and anemia

- All drug-related AEs were Grade 1 or 2 except for:
 - 1 patient with Grade 3 anemia entered study with Grade 2 anemia and had rapid disease progression, receiving only 2 doses of study drug.
 - 1 patient with Grade 4 CRS that resolved quickly with steroids. This patient was noted retrospectively to have subclinical elevated IL-6 serum levels likely due to a case of active gout at baseline.
 - 1 patient with Grade 3 increased lipase that resolved on its own without any clinical symptoms.
 - 5 patients with transient (3-7 days) Grade 3 or 4 lymphopenias that were not associated with adverse outcomes. Transient lymphopenia is a known effect of IL-2 treatment as lymphocytes traffic out of blood and into tissue.

Table 4: Most Common Drug-Related AEs in Phase 1 and 2

Most adverse events were low grade (1 or 2).

Drug-Related AEs in > 5% of Patients n=75		
Adverse Event	Grade 1 or 2 n (%)	Grade 3 or 4 n (%)
Pyrexia	13 (17)	0
Chills	13 (17)	0
Fatigue	10 (13)	0
Nausea	10 (13)	0
Injection Site Reactions	7 (9)	0
Infusion-Related Reactions	7 (9)	0
Lymphopenia	1 (1)	5 (7)
CRS	3 (4)	1 (1)
Anemia	3 (4)	1 (1)
AST Elevation	4 (5)	0
Headache	4 (5)	0

Conclusions

- AU-007 and low-dose, subcutaneous aldesleukin demonstrate a manageable toxicity profile with no sign of vascular leak syndrome or pulmonary edema at all AU-007 and subcutaneous aldesleukin doses evaluated.
- AU-007 PK data demonstrate dose-proportionality over the dose range tested, no signs of neutralizing ADAs activity, and initial T1/2 estimated to be approximately 15 days in humans.
- Peripheral IFN-γ levels and circulating effector cell populations show higher increases with escalating doses of subcutaneous aldesleukin levels, with the greatest benefit achieved at 135K IU/kg of subcutaneous aldesleukin.
- Treg cells decreased in the periphery in the presence of AU-007 (at all dose levels, regardless of subcutaneous aldesleukin dosing regimen). Greater reductions in Tregs were associated with longer PFS. Taken together, the data support the hypothesis that AU-007 can control and redirect the endogenously produced IL-2 and the exogenously administered IL-2 to reduce the Treg population and increase peripheral IFN-γ levels and circulating CD8+ T and NK effector cell populations.
- Dosing with 9 mg/kg of AU-007 was optimal at enhancing IFN-γ production, reducing Tregs, and expanding effector cell populations.
- AU-007 is unique in the IL-2 therapeutic class in its ability to reduce the Treg cell count by binding free IL-2 (endogenous or exogenous) and redirecting it to the dimeric IL-2 receptors expressed on CD8+ T and NK effector cells, expanding those cell populations. This has the effect of biasing the immune system toward activation rather than suppression.
- No clear trends were yet observed in safety or efficacy to determine if a single loading dose (B regimen) or multiple doses of subcutaneous aldesleukin (C regimen) would lead to better outcomes, and clinical investigation to determine the optimal schedule is ongoing in the Phase 2 dose expansion cohorts.
- Based on these results, it was recommended to evaluate 9 mg/kg AU-007 Q2W and 135K IU/kg subcutaneous aldesleukin with both a single loading subcutaneous aldesleukin dose (2B) and Q2W multiple dose subcutaneous aldesleukin (2C) schedules in the Phase 2 expansion cohorts. The Phase 2 expansion is currently ongoing in renal cell carcinoma, melanoma, and non-small cell lung cancer evaluating the 2B and 2C dosing regimens.

Access this presentation via QR code

Presented at the 36th EORTC-NCI-AACR Symposium on Molecular Targets and Cancer Therapeutics, October 23-25, 2024